Aalborg Universitet

Rehabilitation Psychology. A missing link in current Danish practices

Glintborg, Chalotte; Hansen, Tia Gitte Bondesen

Publication date:
2017

Link to publication from Aalborg University

Citation for published version (APA):
Glintborg, C., & Hansen, T. G. B. (2017). *Rehabilitation Psychology. A missing link in current Danish practices*. Poster presented at Annual Convention of the American Psychological Association, Washington D.C., District of Columbia, United States.

General rights
Copyright and moral rights for the publications made accessible in the public portal are retained by the authors and/or other copyright owners and it is a condition of accessing publications that users recognise and abide by the legal requirements associated with these rights.

- Users may download and print one copy of any publication from the public portal for the purpose of private study or research.
- You may not further distribute the material or use it for any profit-making activity or commercial gain
- You may freely distribute the URL identifying the publication in the public portal -

Take down policy
If you believe that this document breaches copyright please contact us at vbn@aub.aau.dk providing details, and we will remove access to the work immediately and investigate your claim.

Downloaded from vbn.aau.dk on: April 25, 2024

REHABILITATION PSYCHOLOGY: A MISSING LINK IN CURRENT DANISH PRACTICES

Glintborg, C. & Hansen, T.G.B (presenter)
Center for Developmental & Applied Psychological Science, Aalborg University, Denmark

Objectives & Methods

OBJECTIVES: Since midst 1990s the psychosocial consequences following an acquired brain injury has been known as the biggest challenge facing rehabilitation. National best practice guidelines in Denmark for individuals suffering from acquired brain injuries recommend coordinated rehabilitation programmes to ensure seamless service transitions and comprehensive rehabilitation practices. However, this recommendation for practice and the evidence informing its advancement are based on unexamined assumptions. Therefore, this study investigated bio-psycho-social outcomes and perceptions of a coordinated rehabilitation programme.

METHOD: In a prospective, naturalistic mixed methods study, rehabilitation outcomes for 82 adults (18-66 years) with moderate to severe acquired brain injury were investigated. Clients who received the coordinated rehabilitation programme KORE (n = 27) were compared to clients from the same area who received standard rehabilitation (SR) prior to the implementation of the KORE programme (n = 37) and clients who received other coordinated efforts (ALT) elsewhere (n = 18). The study employed quantitative data from standardized tests (Functional Independence Measure, Major Depression Inventory, Quality of life, and Impact on Participation and Autonomy Questionnaire) as well as information on return to work and qualitative interviews with clients (N = 82) and their relatives (N = 40).

Results

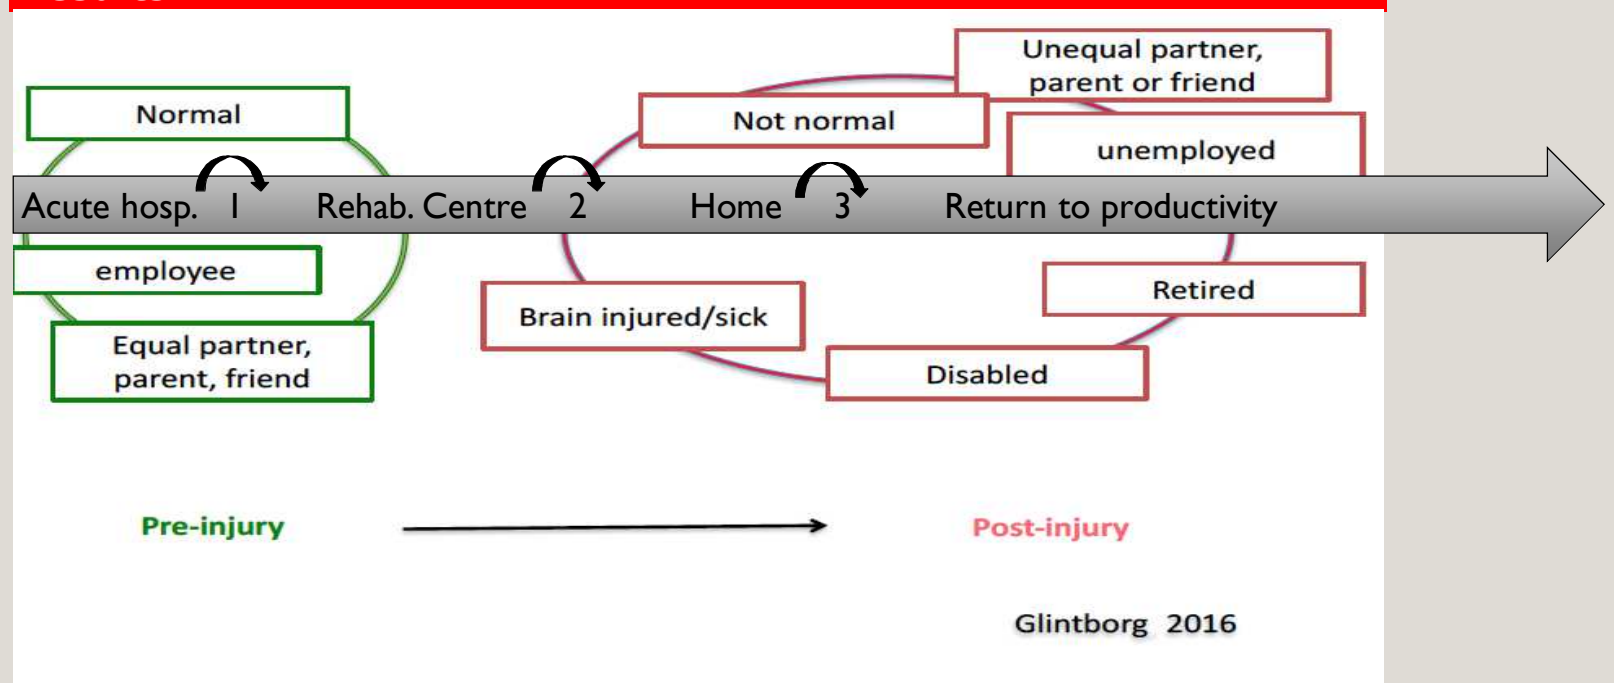
Normal
Not normal
Unequal partner, parent or friend
unemployed
Acute hosp. 1
Rehab. Centre 2
Home 3
Return to productivity
employee
Equal partner, parent, friend
Brain injured/sick
Retired
Disabled
Pre-injury
Post-injury
Glintborg 2016

Results 1: Quan

Functional Independence Measure (FIM) median scores

	SR $n=37$ Median	KORE $n=27$ Median	ALT $n=18$ Median
FIM 0 total	102	97	72[c]
FIM 0 motor	78	71	50[c]
FIM 0 cognitive	25	26	23[c]
FIM 1 total	119[a]	118	109
FIM 1 motor	89[a]	87	80
FIM 1 cognitive	32[a]	31	29
FIM 2 total	119	117[b]	114[a]
FIM 2 motor	87	87[b]	83[a]
FIM 2 cognitive	32	31	32[a]

Note. a = 1 missing value, b = 2 missing values, c = 3 missing values.

QOL mean scores

	SR $n=37$ Mean *(SD)*	KORE $n=27$ Mean *(SD)*	ALT $n=18$ Mean *(SD)*
Time 1 physical		45.3 (20.2)	39.1 (19.1)
Time 1 psychological		55.4 (20.2)	44.0 (18.4)
Time 1 social		71.3 (15,6)	66.0 (20.4)
Time 1 environment		56.6 (13.2)	56.1 (11.5)
Time 2 physical	46.7 (21.7)	47.0 (22.8)[a]	52.8 (22.7)[b]
Time 2 psychological	55.8 (17.9)	53.0 (22.8)[a]	53.2 (20.2)[b]
Time 2 social	69.3 (15.8)	67.3 (22.1)[a]	66.8 (17.1)[b]
Time 2 environment	63.6 (13.6)	56.6 (17.2)[a]	62.7 (12.3)[b]

Note. a = 3 missing values, b = 1 missing value.

Depression (MDI) mean scores

	SR ($n=37$)			KORE ($n=27$)			ALT ($n=18$)		
	min	max	Mean *(SD)*	min	max	Mean *(SD)*	min	max	Mean *(SD)*
MDI time 1				1	47	14.9 (11.1)	5	42	15.5 (10.2)
MDI time 2	1	39	15.4 (9.4)	6	45	18.8 (10.6)[a]	0	43	16.8 (11.2)[b]

Note. a = 4 values missing, b = 1 value missing.

Impact on autonomy and participation (IPAQ) median scores

	SR $n=37$ Median	KORE $n=27$ Median	ALT $n=18$ Median
IPAQ time 1 indoor autonomy		1[a]	2
IPAQ time 1 family roles		1[e]	4[b]
IPAQ time 1 outdoor autonomy		2[c]	3
IPAQ time 1 social relations		0[d]	1[d]
IPAQ time 2 indoor autonomy	0	0[b]	0[a]
IPAQ time 2 family roles	1	2[b]	2[a]
IPAQ time 2 outdoor autonomy	2	2[b]	2[a]
IPAQ time 2 social relations	1	1[b]	1[a]

Note. a = 1 missing value, b = 2 missing values, c = 3 missing values, d = 4 missing values, e = 6 missing values.

Implications/future perspectives

BackUp!

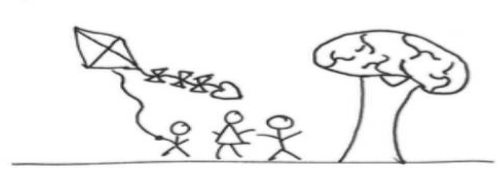

For adults with an acquired brain injury and their close relatives

© Clinic for Handicap og Rehabiliteringspsychology (CHaRe)

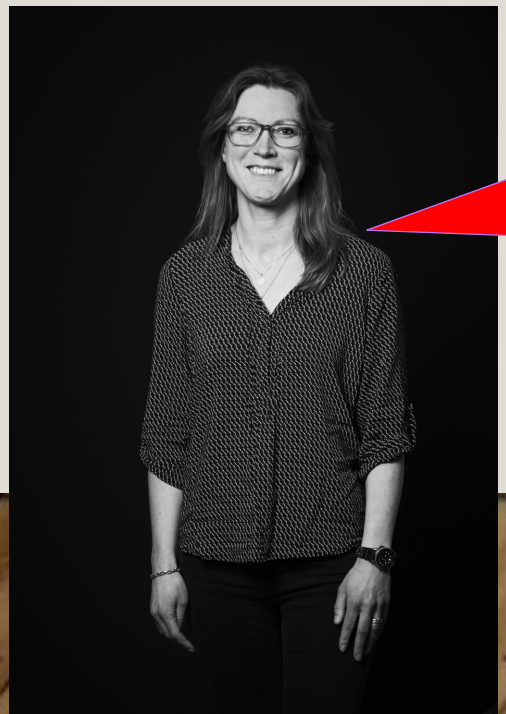

Chalotte Glintborg
Ph.D. Assistant Professor
Department of Communication and Psychology, Center for Developmental & Applied Psychological Science (CeDAPS)
Aalborg University, Kroghstræde 3, 9220 Aalborg Ø, Denmark
Telephone: (+45) 61145665
Email: cgl@hum.aau.dk
Web: www.cedaps.aau.dk

Results 2: Qual

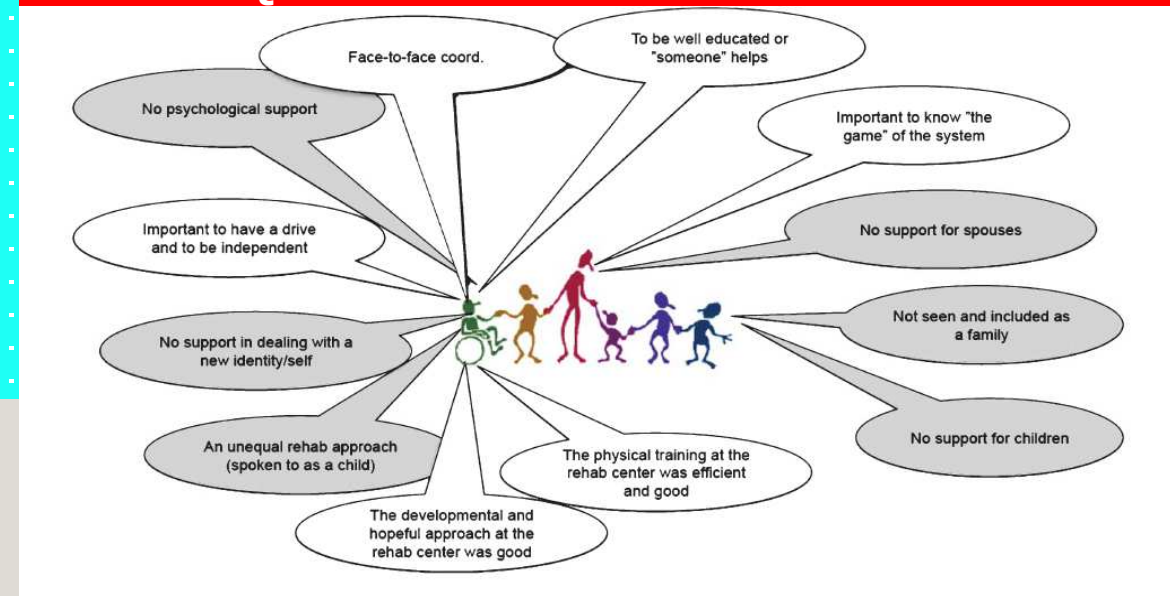
Face-to-face coord.
To be well educated or "someone" helps
No psychological support
Important to know "the game" of the system
Important to have a drive and to be independent
No support for spouses
No support in dealing with a new identity/self
Not seen and included as a family
An unequal rehab approach (spoken to as a child)
No support for children
The physical training at the rehab center was efficient and good
The developmental and hopeful approach at the rehab center was good

What helps and hinders outcome according to interview with clients and relatives.
Note. Green = helpers, Orange = hinderers. (Glintborg & Hansen, 2016)
.

Results, Discussion & Conclusion

RESULTS: Results found no support for the KORE programmes influence on bio-psychosocial outcomes. The clients level of difficulties as measured on FIM, MDI, QOL and IPAQ, and their return to work rate, were at the same level in the group who received the KORE programme as in the group who underwent standard rehabilitation before the KORE programme was implemented. With regard to psychosocial outcomes, one third of all clients showed signs of depression and more than half (50-60%) of all clients experienced dissatisfaction with QOL. Furthermore, 50% of all clients reported problems in their family relations. Qualitative interviews suggested several barriers to improvement, such as unmet needs for psychological support, as rehabilitation services focused on physical and practical training.

DISCUSSION & CONCLUSION: Coordinated interventions do not guarantee comprehensive rehabilitation and better outcomes for clients with acquired brain injury. Psychological support may be the missing link. Rehabilitation psychology is a growing field in European practice that involves applying psychological knowledge to address psychosocial consequences related to individuals with disabilities. Therefore, based on this research, we have developed a new master programme, Clinic for Handicap and Rehabilitation Psychology (CHaRe), at the Department of Communication and Psychology at Aalborg University. CHaRe aims at qualifying students for future work as rehabilitation psychologists. The programme spans 4 semesters and is organized partly in relation to curriculum, partly in relation to clinical practice where students will offer a manual based intervention to clients with disabilities (acquired brain injuries) and their close relatives.

References

Glintborg, C. (2016) Disabled & not normal. Identity construction of adults following an Acquired Brain Injury, *Narrative Inquiry*, 25(1), 1-22.

Glintborg, C. & Hansen, T. (2016). Bio-psycho-social effects of a coordinated neurorehabilitation programme: A naturalistic mixed methods study. *NeuroRehabilitation*, 38, 99–113.

125th ANNUAL CONVENTION | Washington, D.C. AUGUST 3–6, 2017