Aalborg Universitet

Catalogue of EQ-5D Scores for Chronic Conditions in Denmark (poster)

Hvidberg, Michael Falk; Petersen, Karin Dam; Ehlers, Lars Holger

Publication date:
2013

Document Version
Early version, also known as pre-print

Link to publication from Aalborg University

Citation for published version (APA):
Hvidberg, M. F., Petersen, K. D., & Ehlers, L. H. (2013). *Catalogue of EQ-5D Scores for Chronic Conditions in Denmark (poster)*. Poster presented at ISPOR 16th Annual European Congress to be held 2-6 November 2013 at The Convention Centre Dublin in Dublin, Ireland, Dublin, Ireland.

General rights
Copyright and moral rights for the publications made accessible in the public portal are retained by the authors and/or other copyright owners and it is a condition of accessing publications that users recognise and abide by the legal requirements associated with these rights.

- Users may download and print one copy of any publication from the public portal for the purpose of private study or research.
- You may not further distribute the material or use it for any profit-making activity or commercial gain
- You may freely distribute the URL identifying the publication in the public portal -

Take down policy
If you believe that this document breaches copyright please contact us at vbn@aub.aau.dk providing details, and we will remove access to the work immediately and investigate your claim.

Downloaded from vbn.aau.dk on: April 24, 2024

DANISH CENTER FOR
HEALTHCARE IMPROVEMENTS

"CATALOGUE OF EQ-5D SCORES FOR CHRONIC CONDITIONS IN DENMARK"

Michael Falk Hvidberg[1]. Karin Dam Pedersen[1]. Lars H. Ehlers[1] .
[1]Danish Center for Healthcare Improvements, Aalborg University, Denmark.
Corresponding author: hvidberg@business.aau.dk

BACKGROUND:

Chronic patients consume about 70-80% of all healthcare costs in Denmark equivalent to almost $30 billion a year. According to the Danish National Board of Health the numbers are growing (1) - as for many other countries. Even without the on-going economic crisis, the growth in expenditures is not durable in the long term. Thus, there is a need for prioritization if public universal healthcare is to be obtained in the future.

American and English authorities recommend use of standardized methods such as EQ-5D in healthcare evaluations (2,3). Hence, researchers have developed tools for healthcare evaluation in the US and UK - among this the so called EQ-5D score catalogues containing HRQoL for various of chronic conditions (3,4).

In Denmark the EQ-5D lack a systematic – or even widespread – use despite the fact that there al ready has been estimated national Danish preference-based index scores for EQ-5D in 2006 (6) as well as population norms for gender, education and age (7).

A Danish catalogue of EQ-5D scores for chronic conditions is expected to be of great value to Danish regions, municipalities, researchers and other professionals in pursuit of ensuring *quality*, *prioritization* and *optimization* of resources in health care.

OBJECTIVES: EQ-5D catalogues have been developed and tested in US and UK (3,4). The current study aims to develop a Danish preference-based EQ-5D 3L scores catalogue for approximately a hundred of the most common monitored chronic conditions. The development is based on experiences from the US and UK, but adding new factors of importance such as health habits, BMI, social networks and stress.

METHODS: The marginal disutility estimates will be calculated using OLS and CLAD regression on a single source population from a random sample: the National Danish Health Survey from 2010. The survey is a self-administrated survey with approx. 38.557 respondents age ≥16. The survey data is combined with data from national registers containing individual health data e.g. diagnosed chronic conditions during hospitalization, medication, use of hospitals as well as socio-economic data. The catalogue differs from UK and US catalogues' by adding health habits information and by using ICD-10 classifications from registers as well as it is based on Danish EQ-5D tariffs. The marginal disutility — as well as unadjusted mean and medians — is calculated for each chronic condition controlling for age, gender, ethnicity, family income, education and comorbidity etc.

RESULTS: Marginal disutility estimates (EQ-5D) for approximately a hundred ICD-10 chronic conditions are planned to be estimated and compared (however, 21 selected conditions are illustrated here). It is expected that this new knowledge will contribute and qualify prioritization debate, when results are published and combined with knowledge of for example factors of importance and burden of disease in costs.

PRELIMINARY RESULTS FOR CHOOSEN CONDITIONS & HEALTH HABITS:

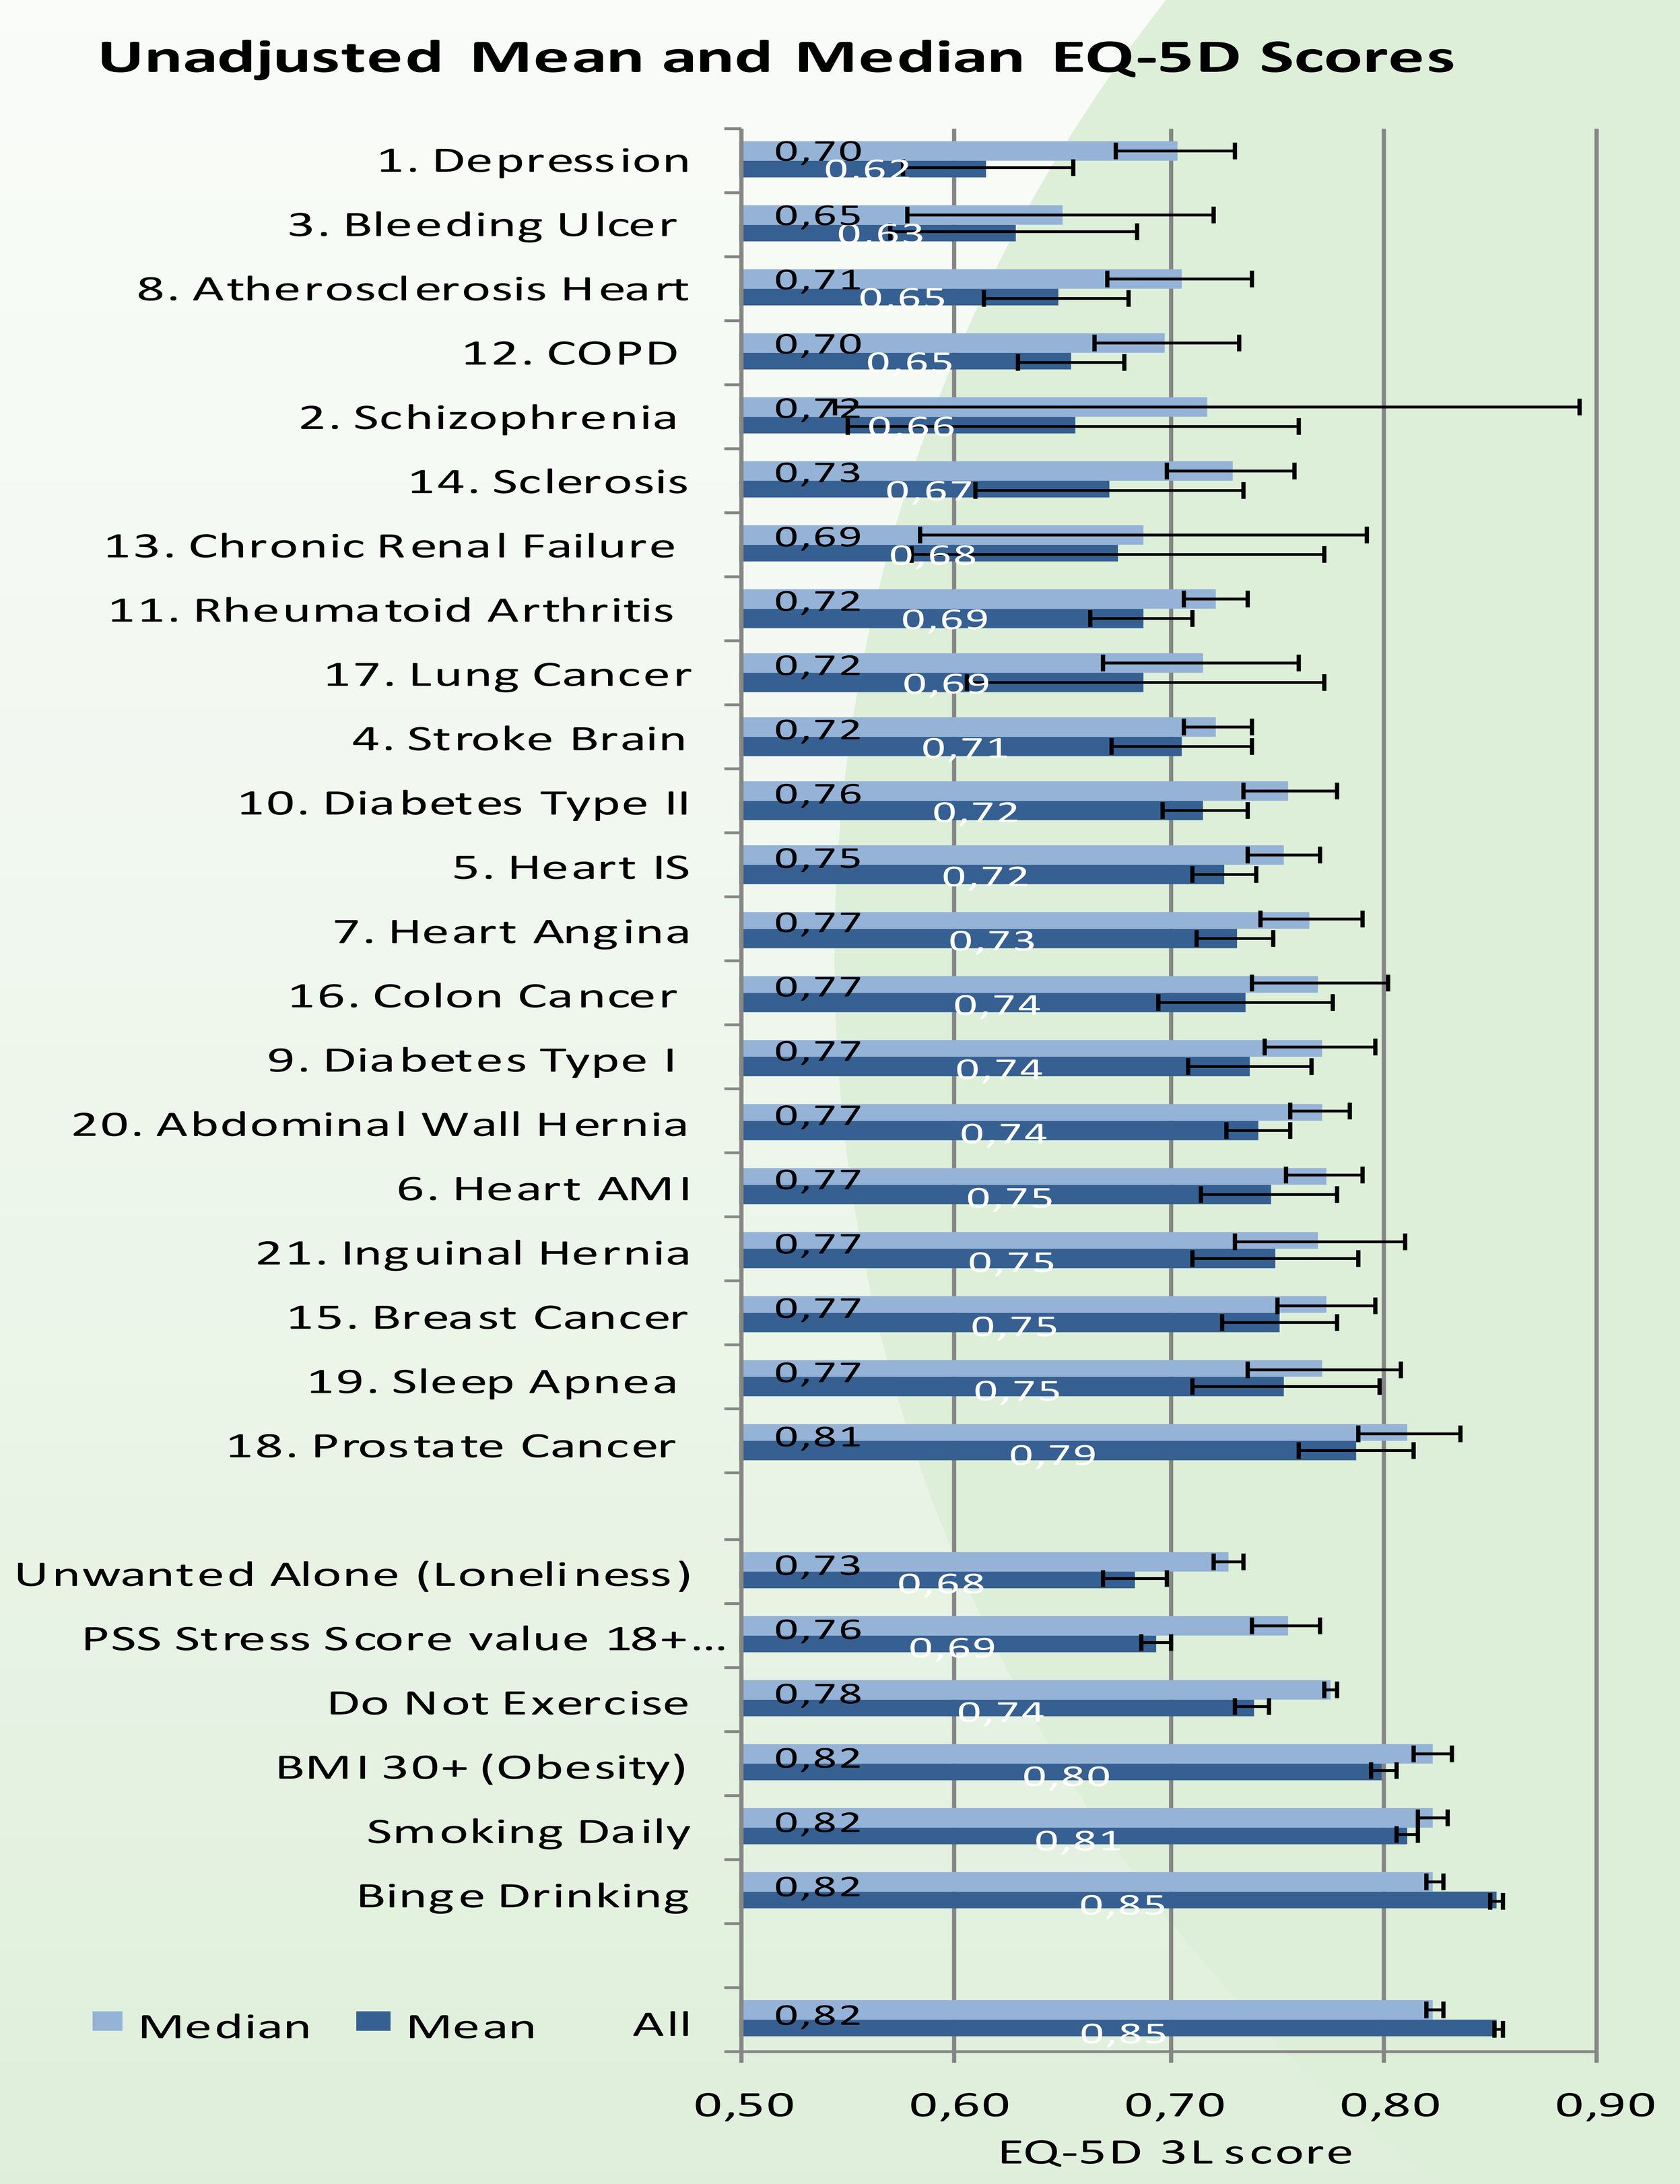
Unadjusted Mean and Median EQ-5D Scores
1. Depression 0,70 0.62
3. Bleeding Ulcer 0,65 0.63
8. Atherosclerosis Heart 0,71 0.65
12. COPD 0,70 0.65
2. Schizophrenia 0,72 0.66
14. Sclerosis 0,73 0.67
13. Chronic Renal Failure 0,69 0.68
11. Rheumatoid Arthritis 0,72 0.69
17. Lung Cancer 0,72 0.69
4. Stroke Brain 0,72 0.71
10. Diabetes Type II 0,76 0.72
5. Heart IS 0,75 0.72
7. Heart Angina 0,77 0.73
16. Colon Cancer 0,77 0.74
9. Diabetes Type I 0,77 0.74
20. Abdominal Wall Hernia 0,77 0.74
6. Heart AMI 0,77 0.75
21. Inguinal Hernia 0,77 0.75
15. Breast Cancer 0,77 0.75
19. Sleep Apnea 0,77 0.75
18. Prostate Cancer 0,81 0.79
Unwanted Alone (Loneliness) 0,73 0.68
PSS Stress Score value 18+... 0,76 0.69
Do Not Exercise 0,78 0.74
BMI 30+ (Obesity) 0,82 0.80
Smoking Daily 0,82 0.81
Binge Drinking 0,82 0.85
All 0,82 0.85
Median
Mean
0,50 0,60 0,70 0,80 0,90
EQ-5D 3L score

OLS regression on EQ-5D (ICD-10 codes in brackets)	**Coef.**	**Robust std. Err.**
1. Depression (F32 F33 F34.1 F06.32)	-0,113**	0,017
2. Schizophrenia (F20)	-0,066	0,041
3. Bleeding Ulcer (K250 K254 K260 K264 K270 K274)	-0,028	0,022
4. Stroke Brain (I63)	-0,043**	0,014
5. Heart IS (I20 I21 I22 I23 I24 I25)	-0,041**	0,012
6. Heart AMI (I21 I22)	0,028*	0,015
7. Heart Angina (I20)	-0,014	0,013
8. Atherosclerosis Heart (I110 I130 I132 I420 I426 I427 I429 I500 I501 I509)	-0,047**	0,016
9. Diabetes Type I (E10)	-0,041**	0,011
10. Diabetes Type II (E11)	-0,032**	0,009
11. Rheumatoid Arthritis (M05 M06 M07 M10 M11 M12 M13 M14)	-0,104**	0,011
12. COPD (J96 J44 J96 J13 J14 J15 J16 J17 J18)	-0,088**	0,012
13. Chronic Renal Failure (I120 I131 I132 N17)	-0,054	0,038
14. Sclerosis (G35)	-0,120**	0,025
15. Breast Cancer (C50)	-0,043**	0,012
16. Colon Cancer (C18)	-0,027	0,016
17. Lung Cancer (C34)	-0,078*	0,043
18. Prostate Cancer (C61)	-0,011	0,011
19. Sleep Apnea (G473)	-0,031*	0,017
20. Abdominal Wall hernia (K40)	-0,010	0,019
21. Inguinal Hernia (K43)	-0,018**	0,008
BMI 30+ (Obesity)	-0,033**	0,003
Smoking Daily	-0,021**	0,002
Binge Drinking	-0,002	0,003
Do Not Exercise	-0,066**	0,003
PSS Stress Score value 18+ (20% most stressed)	-0,155**	0,003
Unwanted Alone (Loneliness)	-0,081**	0,007

*** sig. < 0,01 * sig. < 0,05*
Model controlled for gender, age, education, family income, geography and etnicity. n=36298

REFERENCES:

1. Danish National Board of Health. Kronisk sygdom - Patient, sundhedsvæsen og samfund. Forudsætninger for det gode forløb [Internet]. www.sst.dk; 2005.
2. National Institute of Clinical Excellence (NICE). Social Value Judgements: Principles For The Development Of NICE Guidance. [Internet]. 2008.
3. Sullivan PW, Ghushchyan V. Preference-Based EQ-5D index scores for chronic conditions in the United States. Medical Decision Making [Internet]. 2006 [cited 2012 Apr 23];26(4):410–20.
4. Sullivan PW, Slejko JF, Sculpher MJ, Ghushchyan V. Catalogue of EQ-5D scores for the United Kingdom. Medical Decision Making [Internet]. 2011 [cited 2012 Jun 4];31(6):800–4.
5. Sullivan, Patrick W; Ghushchyan V. How to Use the Estimates [Internet]. 2006. Report, appendix.
6. Wittrup-Jensen KU, Lauridsen J, Gudex C, Pedersen KM. Generation of a Danish TTO value set for EQ-5D health states. Scandinavian Journal of Public Health [Internet]. 2009 Jul [cited 2012 Apr 22];37(5):459–66.
7. Sørensen J, Davidsen M, Gudex C, Pedersen KM, Brønnum-Hansen H. Danish EQ-5D Population Norms. Scandinavian Journal of Public Health [Internet]. 2009 Jul [cited 2012 Mar 23];37(5):467–74.

NORTH DENMARK REGION
AALBORG UNIVERSITY
DENMARK