Aalborg Universitet

Co-creation of Patient Engagement Quality Guidance and Quality Criteria for Medicines Development

An International Multi-stakeholder Initiative

Deane, K; Skovlund, Søren Eik; Nafria, B; Delbecque, L; Gorbenko, OV; Pakarinen, C; Brooke, N; Hamoir, AM; PFMD Patient Engagement Meta-framework Co-creation Team; Skovlund, Søren Eik

Publication date:
2018

Document Version
Publisher's PDF, also known as Version of record

Link to publication from Aalborg University

Citation for published version (APA):
Deane, K., Skovlund, S. E., Nafria, B., Delbecque, L., Gorbenko, OV., Pakarinen, C., Brooke, N., Hamoir, AM., PFMD Patient Engagement Meta-framework Co-creation Team, & Skovlund, S. E. (2018). *Co-creation of Patient Engagement Quality Guidance and Quality Criteria for Medicines Development: An International Multi-stakeholder Initiative*. Poster presented at ISPOR 2018, Spain.

General rights
Copyright and moral rights for the publications made accessible in the public portal are retained by the authors and/or other copyright owners and it is a condition of accessing publications that users recognise and abide by the legal requirements associated with these rights.

- Users may download and print one copy of any publication from the public portal for the purpose of private study or research.
- You may not further distribute the material or use it for any profit-making activity or commercial gain
- You may freely distribute the URL identifying the publication in the public portal -

Take down policy
If you believe that this document breaches copyright please contact us at vbn@aub.aau.dk providing details, and we will remove access to the work immediately and investigate your claim.

Downloaded from vbn.aau.dk on: October 28, 2024

Co-creation of Patient Engagement Quality Guidance and Quality Criteria for Medicines Development: An International Multi-stakeholder Initiative

Deane K[1], Skovlund SE[2], Nafria B[3], Delbecque L[4], Gorbenko OV[5], Pakarinen C[6], Brooke N[6], Hamoir AM[6] on behalf of the PFMD Patient Engagement Meta-framework Co-creation Team (www.pfmd.org)

[1]University of East Anglia, Norwich, UK; [2]Aalborg University, Aalborg, Denmark; [3]Sant Joan de Déu Research Foundation, Santa Rosa, Spain; [4]Pharmerit, International, AV Rotterdam, Belgium; [5]ViiV Healthcare, Middlesex, UK; [6]Patient Focused Medicines Development (PFMD), The Synergist, Brussels, Belgium

PATIENT FOCUSED MEDICINES DEVELOPMENT

BACKGROUND

- The value and necessity of engaging patients during the development and lifecycle of medicines is increasingly recognised and many discrete initiatives are underway or planned.
- Though complementary, these separate activities have resulted in a fragmented patient engagement (PE) landscape. Investment in a holistic, integrated and sustainable initiative is required to cover the entire medicines lifecycle and to connect stakeholders across geographies.
- Patient Focused Medicines Development (PFMD; www.pfmd.org) is a global, multi-stakeholder collaboration of health stakeholders taking a rational four-step approach towards co-creation of a meta-framework for PE: (1) mapping and connecting the PE landscape to learn from existing efforts; (2) multi-stakeholder co-creation of PE guidance and good practice identification; (3) development and piloting of a strawman meta-framework for PE; and (4) refinement of the strawman meta-framework to create an actionable meta-framework and implementation tools.
- This poster focuses on the development of the PE Quality Guidance and identification of good practice examples.

METHODS

PE Quality Guidance co-creation

- Multi-stakeholder working groups (WGs) and task forces (TFs) were established for Guidance co-creation. Published literature and existing frameworks were reviewed (2011-2016). SYNaPse (a PE mapping and networking platform developed by PFMD) was used to identify unpublished PE initiatives and initiative owners were invited to WGs/TFs.
- A preliminary PE Quality Guidance tool (incorporating PE Quality Criteria) was developed by WGs/TFs in an iterative process, through: review of published material and landscape analysis to identify unmet needs; incorporation of participants' PE experience to identify and prioritise needed actions; and owners retrospectively applying the draft Guidance to their initiatives as a pressure-testing evaluation **(Figure 1)**.

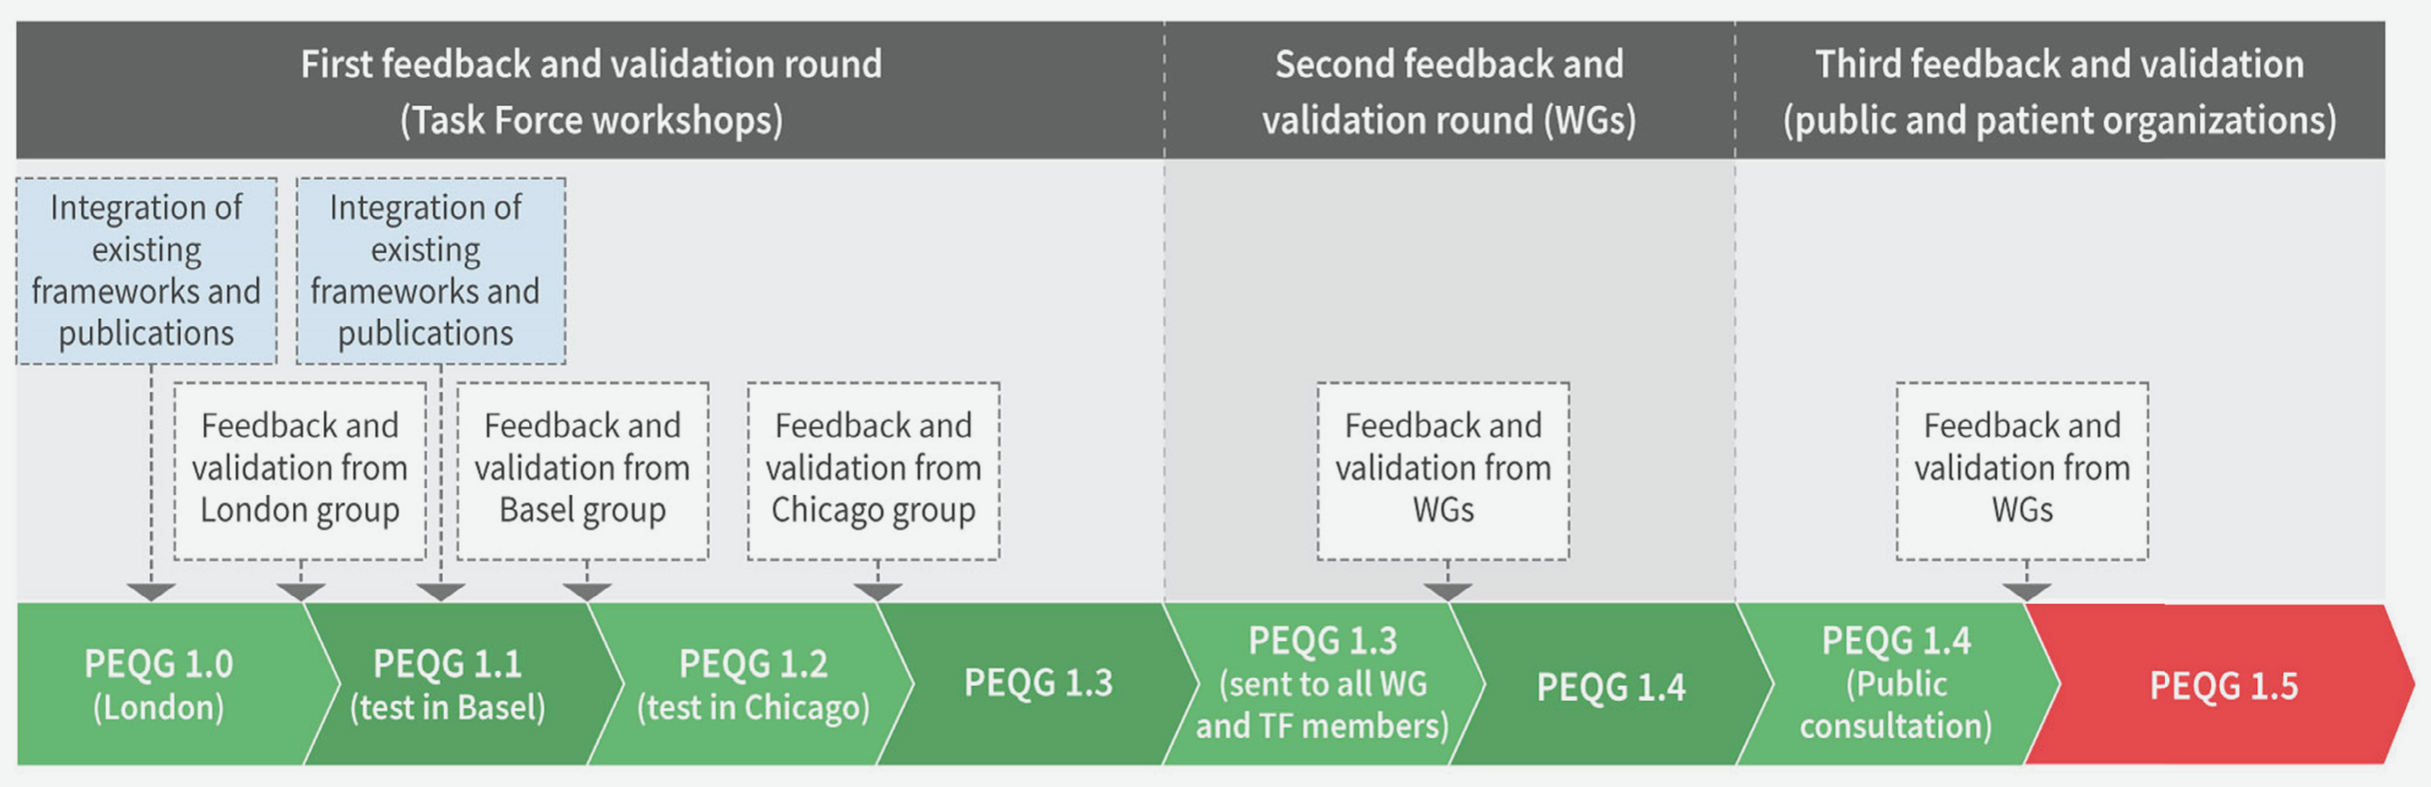
First feedback and validation round (Task Force workshops)
Second feedback and validation round (WGs)
Third feedback and validation (public and patient organizations)
Integration of existing frameworks and publications
Integration of existing frameworks and publications
Feedback and validation from London group
Feedback and validation from Basel group
Feedback and validation from Chicago group
Feedback and validation from WGs
Feedback and validation from WGs
PEQG 1.0 (London)
PEQG 1.1 (test in Basel)
PEQG 1.2 (test in Chicago)
PEQG 1.3
PEQG 1.3 (sent to all WG and TF members)
PEQG 1.4
PEQG 1.4 (Public consultation)
PEQG 1.5

Figure 1: Process for development of the PE Quality Guidance (PEQG) tool

Public consultation on draft PE Quality Guidance

- A public online, survey-based consultation on the draft PE Quality Guidance was undertaken (from 20 November 2017 to 1 January 2018) to gather wider input from PE stakeholders.
- Consultation was invited via the PFMD website and across stakeholder groups via WG, TF and PFMD networks; and through advertising (21,589 reach), media (40,259 reach) and a social media campaign. Feedback from the public consultation and from a separate focus group was used to refine the PE Quality Guidance.

Identification of good practice examples for the Book of Good Practice (BOGP)

Examples of good practice were collected through screening of initiatives in SYNaPsE and via TF and PFMD networks. The review and selection process for inclusion in the BOGP was anonymised to avoid bias and was undertaken by a multi-stakeholder PE Meta-framework Core Team.

RESULTS: PE QUALITY GUIDANCE CO-CREATION

Co-creation workshops and participants

- Nine WG/TF/Core Team meetings were held (Nov 2016-June 2018) involving 76 unique participants, representing 51 organisations (including patient charities, academic researchers, funders, pharmaceutical companies).

Mapping and connecting the PE landscape

- A total of 239 publications were identified covering the period of 2011-2016 using PE-relevant search criteria (in PubMed and British Medical Journal Open), of which 25 were relevant or somewhat relevant. An additional 8 resources (including seven conceptual frameworks/models[1-10]) were included to capture approaches outside of the search period.

Multi-stakeholder co-creation of PE Quality Guidance

- WGs/TFs adapted and augmented the characteristics from the frameworks into the PE Quality Guidance. The PE Quality Guidance is organised around seven PE Quality Criteria that represent key elements of quality PE. These criteria have drawn primarily from guidance and frameworks developed by INVOLVE[8-10] and enriched with the analysis of PE initiatives collected in SYNaPsE and from the WG/TF/Core Team.

Structure of the PE Quality Guidance

- The PE Quality Guidance is structured to be used either as an aid to planning new PE projects, or to facilitate assessment of ongoing or completed PE projects. It incorporates four components to capture: background information, quality of PE, outcomes and lessons learned.
- The PE Quality Guidance proposes seven PE Quality Criteria **(Table 1)** to aid assessment of the quality of PE in specific projects.
 - The PE Quality Criteria: describe core values that a good PE practice should consider including in its processes; provide a set of principles to improve consistency in PE practices; and enable systematic assessment and communication of project outcomes.
 - For each criterion, there is a definition, a rationale and questions for consideration by the initiative owners that can be used for planning and/or evaluation purposes. The relevance of each PE Quality Criterion may differ from project to project and this variation can be captured within the tool.

Table 1: Overview of the PE Quality Criteria

Quality Criterion	Explanation
1. Shared purpose	This refers to the project's aims and outcomes that all stakeholders taking part should agree on before starting the project. It can facilitate open exchange of views, and help define the scope and objectives of the project.
2. Respect and accessibility	This refers to (1) respecting each other, and respectful interactions within the project to be established among partners, and (2) openness to and inclusion of individuals and communities (to the project) without discrimination. It should consider practical ways of enabling different stakeholders with different needs to take part in the project
3. Representativeness of stakeholders	This refers to the mix of people you involve, which should reflect the needs of the project, and the interests of those who may benefit from project outputs (for example, target population). It should consider diversity in expertise, experience, demographics, and other relevant criteria for inclusion.
4. Roles and responsibilities	This refers to the need for agreed – and ideally co-created – roles and responsibilities, in writing. It should indicate that all aspects of project needs will be established upfront and revisited regularly.
5.Capacity and capability for engagement	This refers to (1) capacity as having relevant and dedicated resources from all stakeholders (e.g., stakeholders allocating sufficient time to allow genuine engagement); and (2) capabilities required for all stakeholders to enable meaningful engagement (e.g., the required level of knowledge or expertise and training).
6. Transparency in communication and documentation	This refers to the establishment of a communications plan and ongoing project documentation that can be shared with stakeholders. It should consider timeliness of communication, project updates to share progress, and how outcomes will be shared to show how contribution of stakeholders was of value to the success of the project.
7. Continuity and sustainability	This refers to the smooth progression of the project, as well as efforts to maintain ongoing relationships with stakeholders. It should consider the role of stakeholders beyond a single project.

Using the PE Quality Guidance

- The PE Quality Guidance contains tools for assessment of initiatives that are either planned or in preparation (scenario 1) or ongoing or completed (scenario 2) against the PE Quality Criteria. The tools can be accessed directly from the PE Quality Guidance and completed online **(Figure 2)**.
- For each scenario, the tool captures basic background information and then focuses on each of the PE Quality Criteria.
- Each criteria section has
 - A detailed explanation of the criterion
 - Supporting questions to help identify specific actions
 - Illustrative examples of that criterion in practice
 - A link to relevant resources
- The section on PE Quality Criteria is followed by the '*Results and outcomes*' section. A final section on *'Lessons learned'* is available in the tool for ongoing or completed projects **(Table 2)**.

Figure 2: Format of the PE Quality Guidance tools

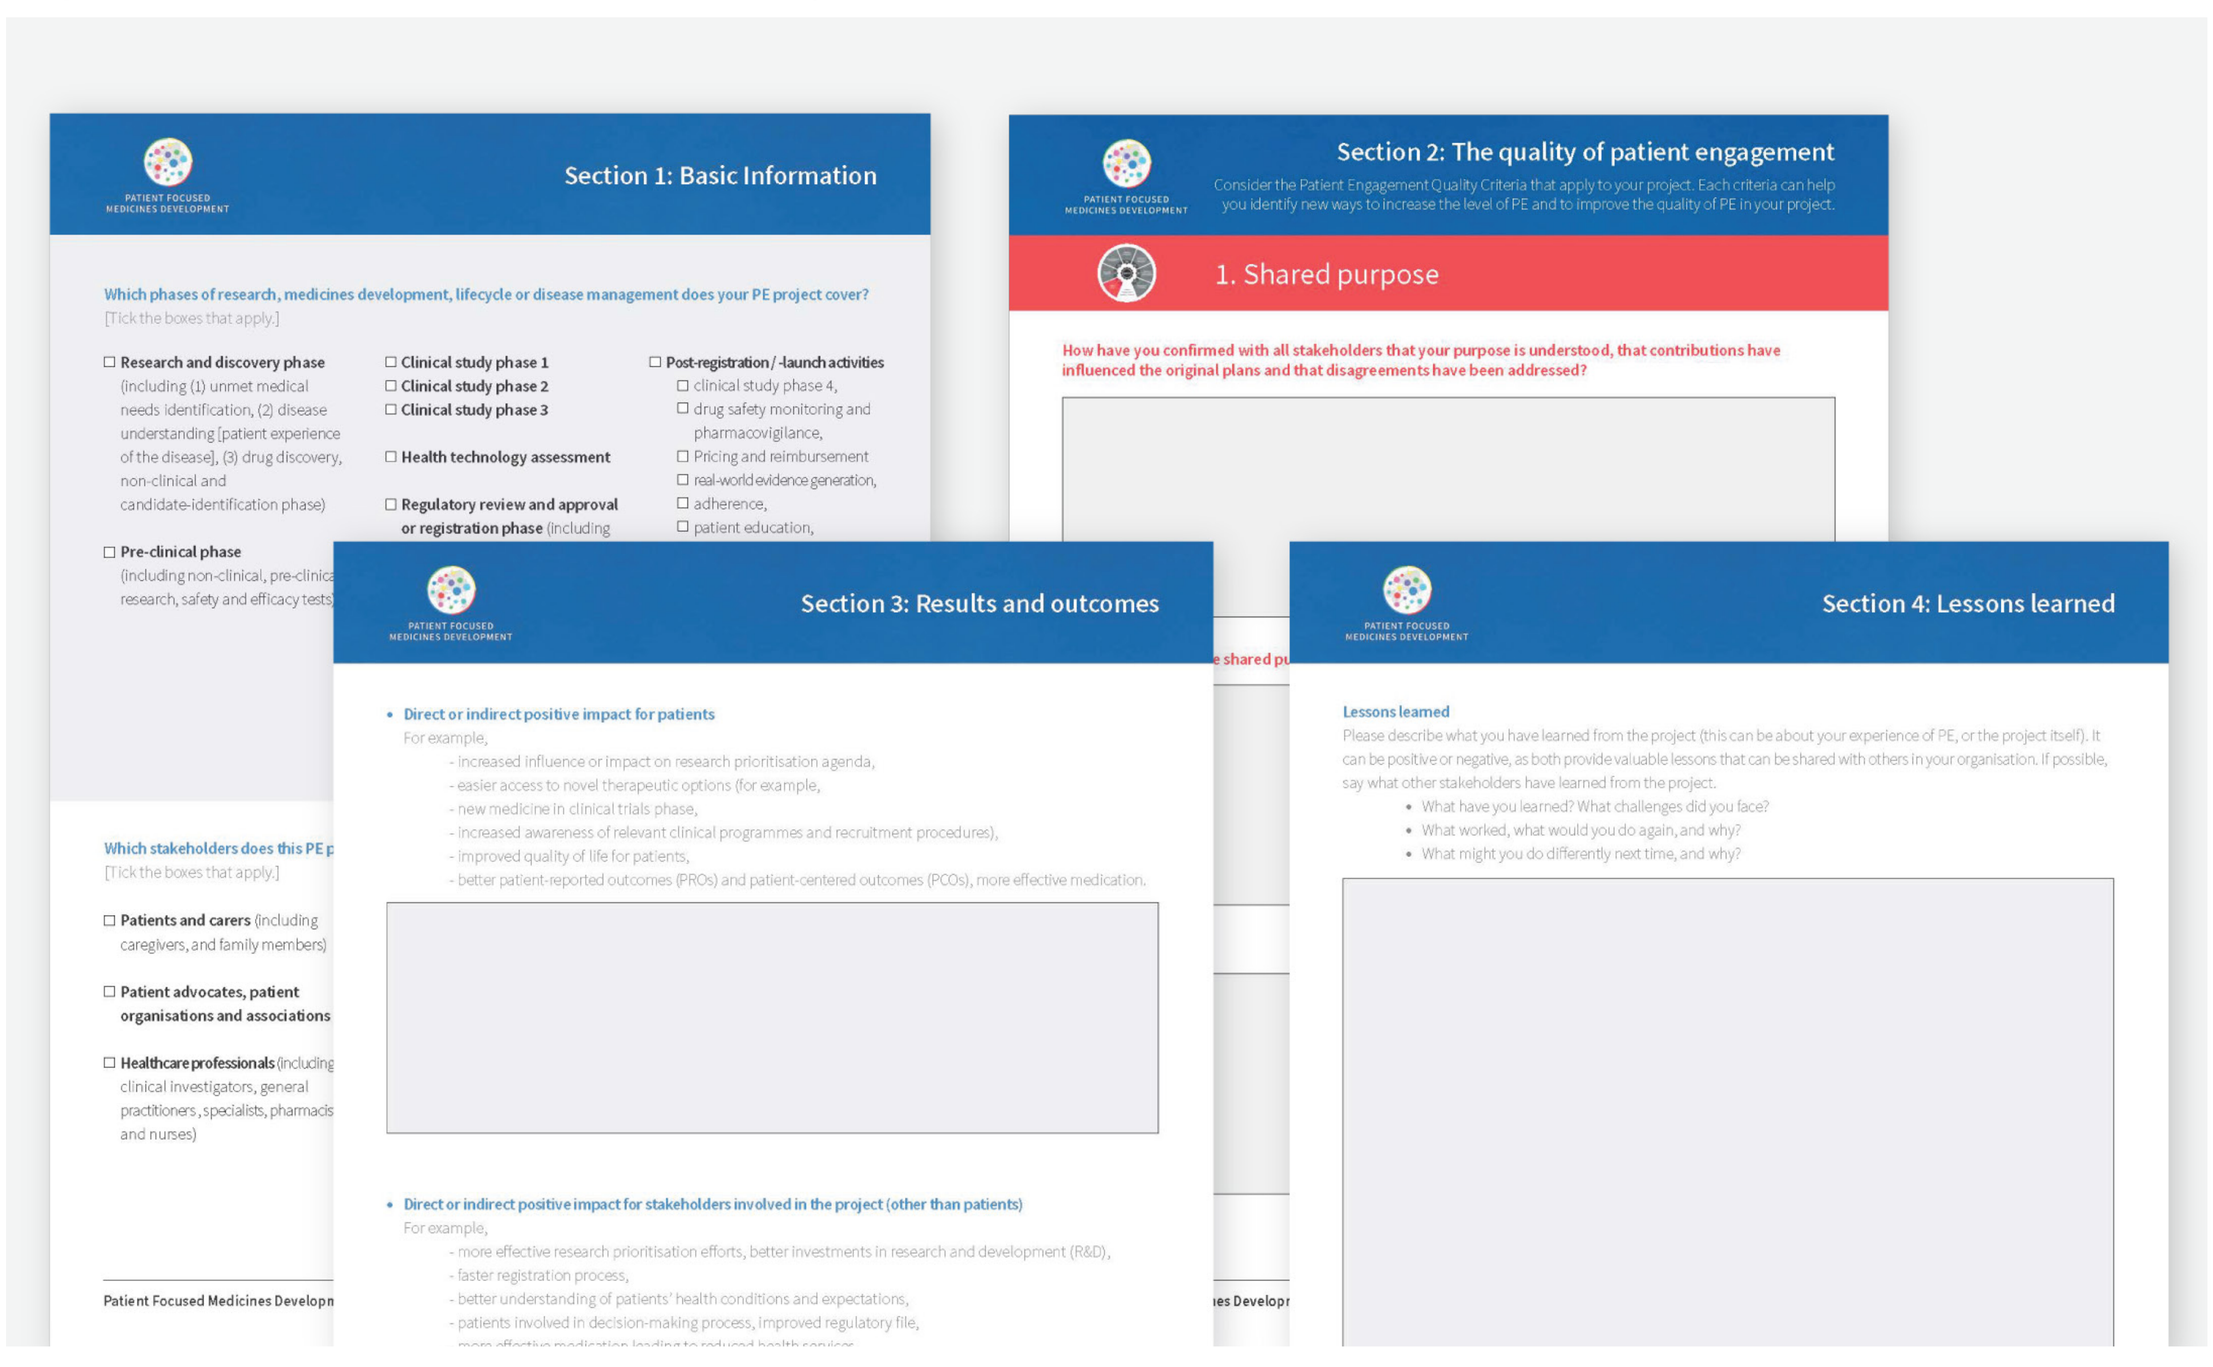
Section 1: Basic Information
Section 2: The quality of patient engagement
1. Shared purpose
Section 3: Results and outcomes
Section 4: Lessons learned

Table 2: Using the PE Quality Guidance tool – step-by-step

Section	Use this section to...
Section 1: Basic information	▪ Describe the project objectives, desired outcomes and methods used ▪ Identify which phases of medicines development, lifecycle or disease management the project covers ▪ Describe which stakeholders the project involves
Section 2: Quality of PE	▪ Systematically 'walk through' each PE project, identifying and detailing specific tactics or approaches to achieve project aims
Section 3: Results and outcomes	▪ Capture expected or desired outcomes for planned projects ▪ Capture actual results and details of methods used to collect them for ongoing or completed projects
Section 4: Lessons learned	▪ Capture experiences from the PE initiative including challenges faces and solutions implemented ▪ Document what went well and what you would do differently in future projects

RESULTS: PUBLIC CONSULTATION ON DRAFT PE QUALITY GUIDANCE

- The public consultation on the draft PE Quality Guidance resulted in 851 website visits and 67 responses (74% completion rate) across diverse stakeholders including: patients/patient organisations; pharma/biotech; and research/academia.
- The majority (69%) of responders were from Europe. Over 80% of respondents indicated that they were 'advanced' in terms of their PE experience ('actively part of PE projects').
- Of 51 respondents, the vast majority (range 85% to 96%) agreed or strongly agreed that: the PE Quality Criteria are useful for achieving quality PE practice; the language used is comprehensive and easy to understand; and the format is clear **(Figure 3)**.
- Specific feedback included needing practical examples and illustrative tips based on how others have used it. The Book of Good Practice (BOGP) was created in response to this feedback
 - The BOGP provides real-life, practical examples of PE projects that are exemplary in one or more PE Quality Criteria or overall show meaningful ways to engage and involve patients and other stakeholders in the medicines research and development continuum
 - All cases included in the BOGP have been anonymised, and the language and content reflects the views of project owners
- Most respondents (range 65% to 84%) agreed or strongly agreed that the PE Quality Guidance could be used to: improve the quality and consistency of PE activities; better plan and develop PE activities; better assess the quality and impact of PE initiatives; identify gaps and opportunities for PE activities; capture and share learnings beyond the project or project team; and, structure work with partnering organisations **(Figure 4)**. The feedback from the public consultation has informed the final versions of the PE Quality Guidance.

Figure 3: General opinion on PE Quality Guidance

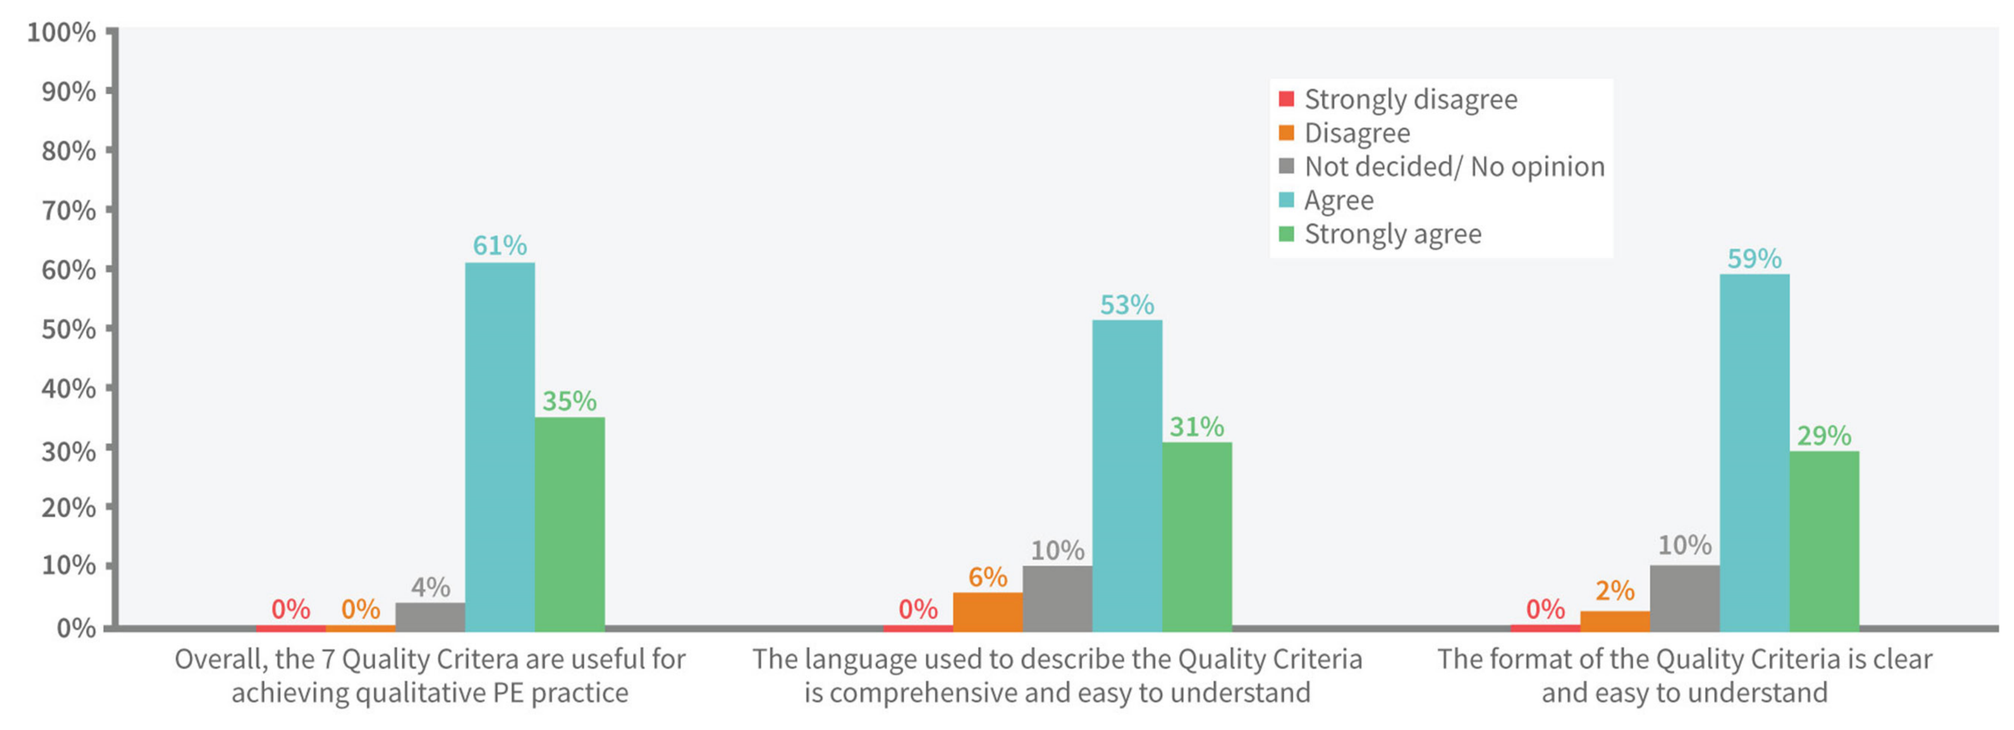
Strongly disagree
Disagree
Not decided/ No opinion
Agree
Strongly agree
Overall, the 7 Quality Critera are useful for achieving qualitative PE practice
0% 0% 4% 61% 35%
The language used to describe the Quality Criteria is comprehensive and easy to understand
0% 6% 10% 53% 31%
The format of the Quality Criteria is clear and easy to understand
0% 2% 10% 59% 29%

Figure 4: Usability of PE Quality Guidance

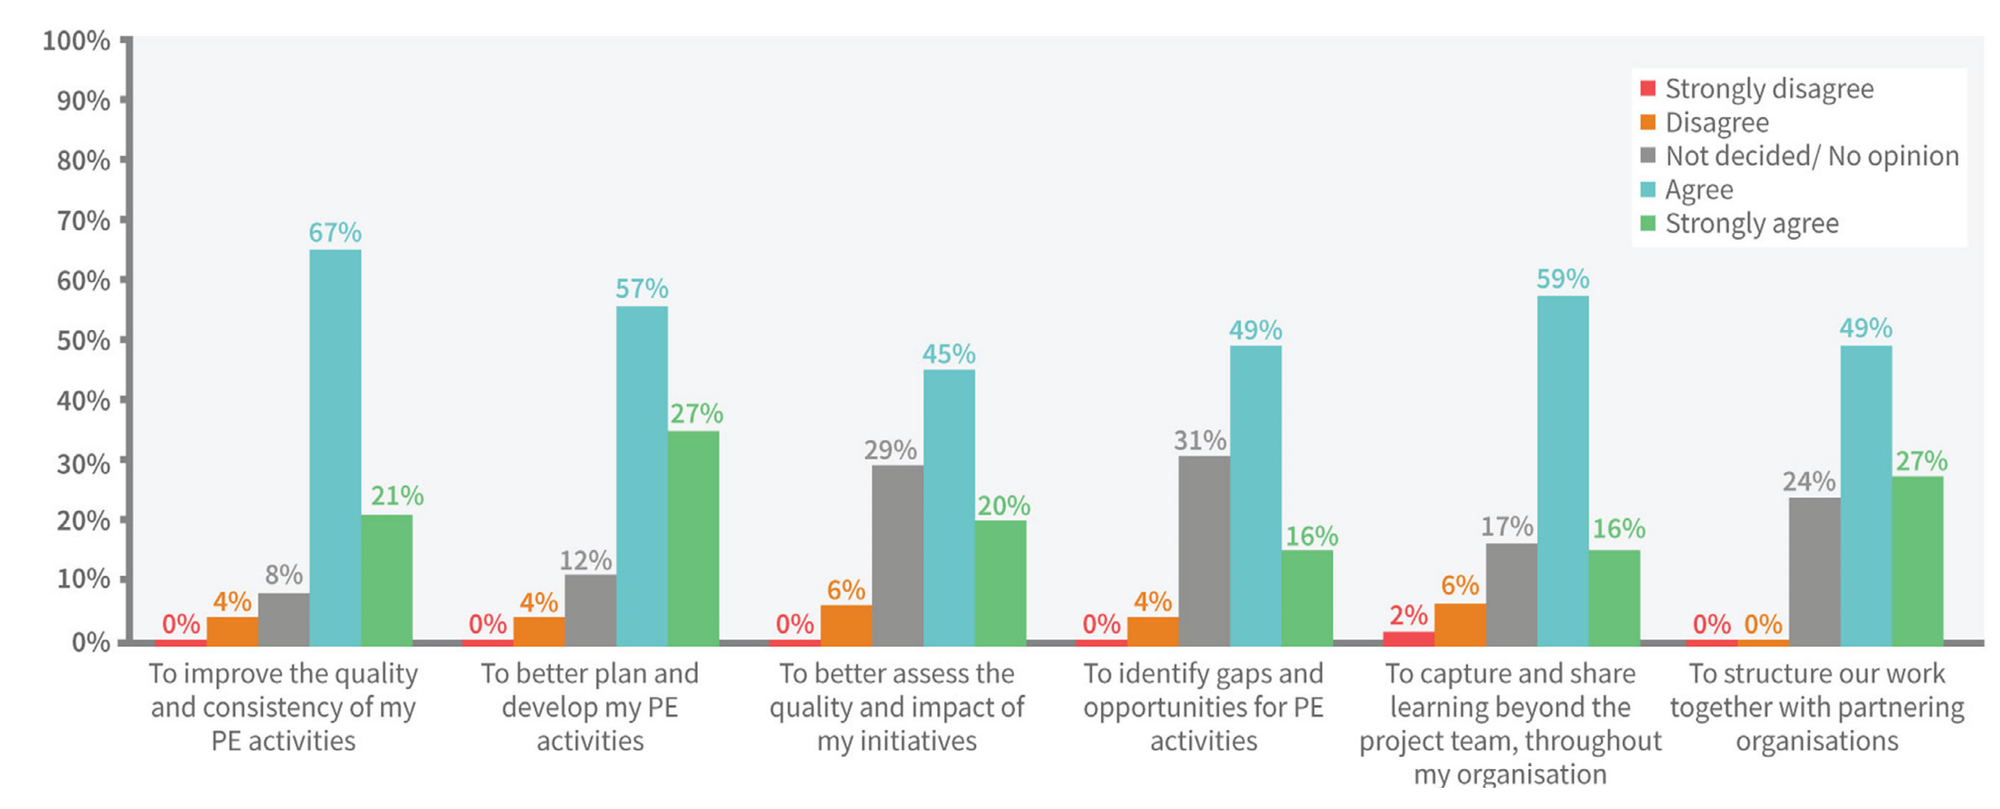
Strongly disagree
Disagree
Not decided/ No opinion
Agree
Strongly agree
To improve the quality and consistency of my PE activities
0% 4% 8% 67% 21%
To better plan and develop my PE activities
0% 4% 12% 57% 27%
To better assess the quality and impact of my initiatives
0% 6% 29% 45% 20%
To identify gaps and opportunities for PE activities
0% 4% 31% 49% 16%
To capture and share learning beyond the project team, throughout my organisation
2% 6% 17% 59% 16%
To structure our work together with partnering organisations
0% 0% 24% 49% 27%

CONCLUSIONS

- The co-created PE Quality Guidance tool can be used by multiple stakeholder groups and provides a practical guide to improve the quality of PE during planning and development of new projects or assess the quality and impact of ongoing/completed projects. The good practice examples provided will support implementation of PE Quality Guidance.
- PE is not 'one size fits all' and consequently, the PE Quality Guidance is not prescriptive, rather it is based on core principles that can be adapted and applied according to the unique needs of each interaction and project.
- We urge use of, and feedback on, the PEQG by diverse stakeholders to drive improvements in PE and allow continuous refinement of the tool.
- The PE Quality Guidance is not an isolated tool. It is part of a wider and ongoing effort towards synergising PE activities for more effective, systematic and measurable PE through co-creation and implementation of a meta-framework for PE **(Figure 5)**.

Figure 5: Roadmap to the PE meta-framework

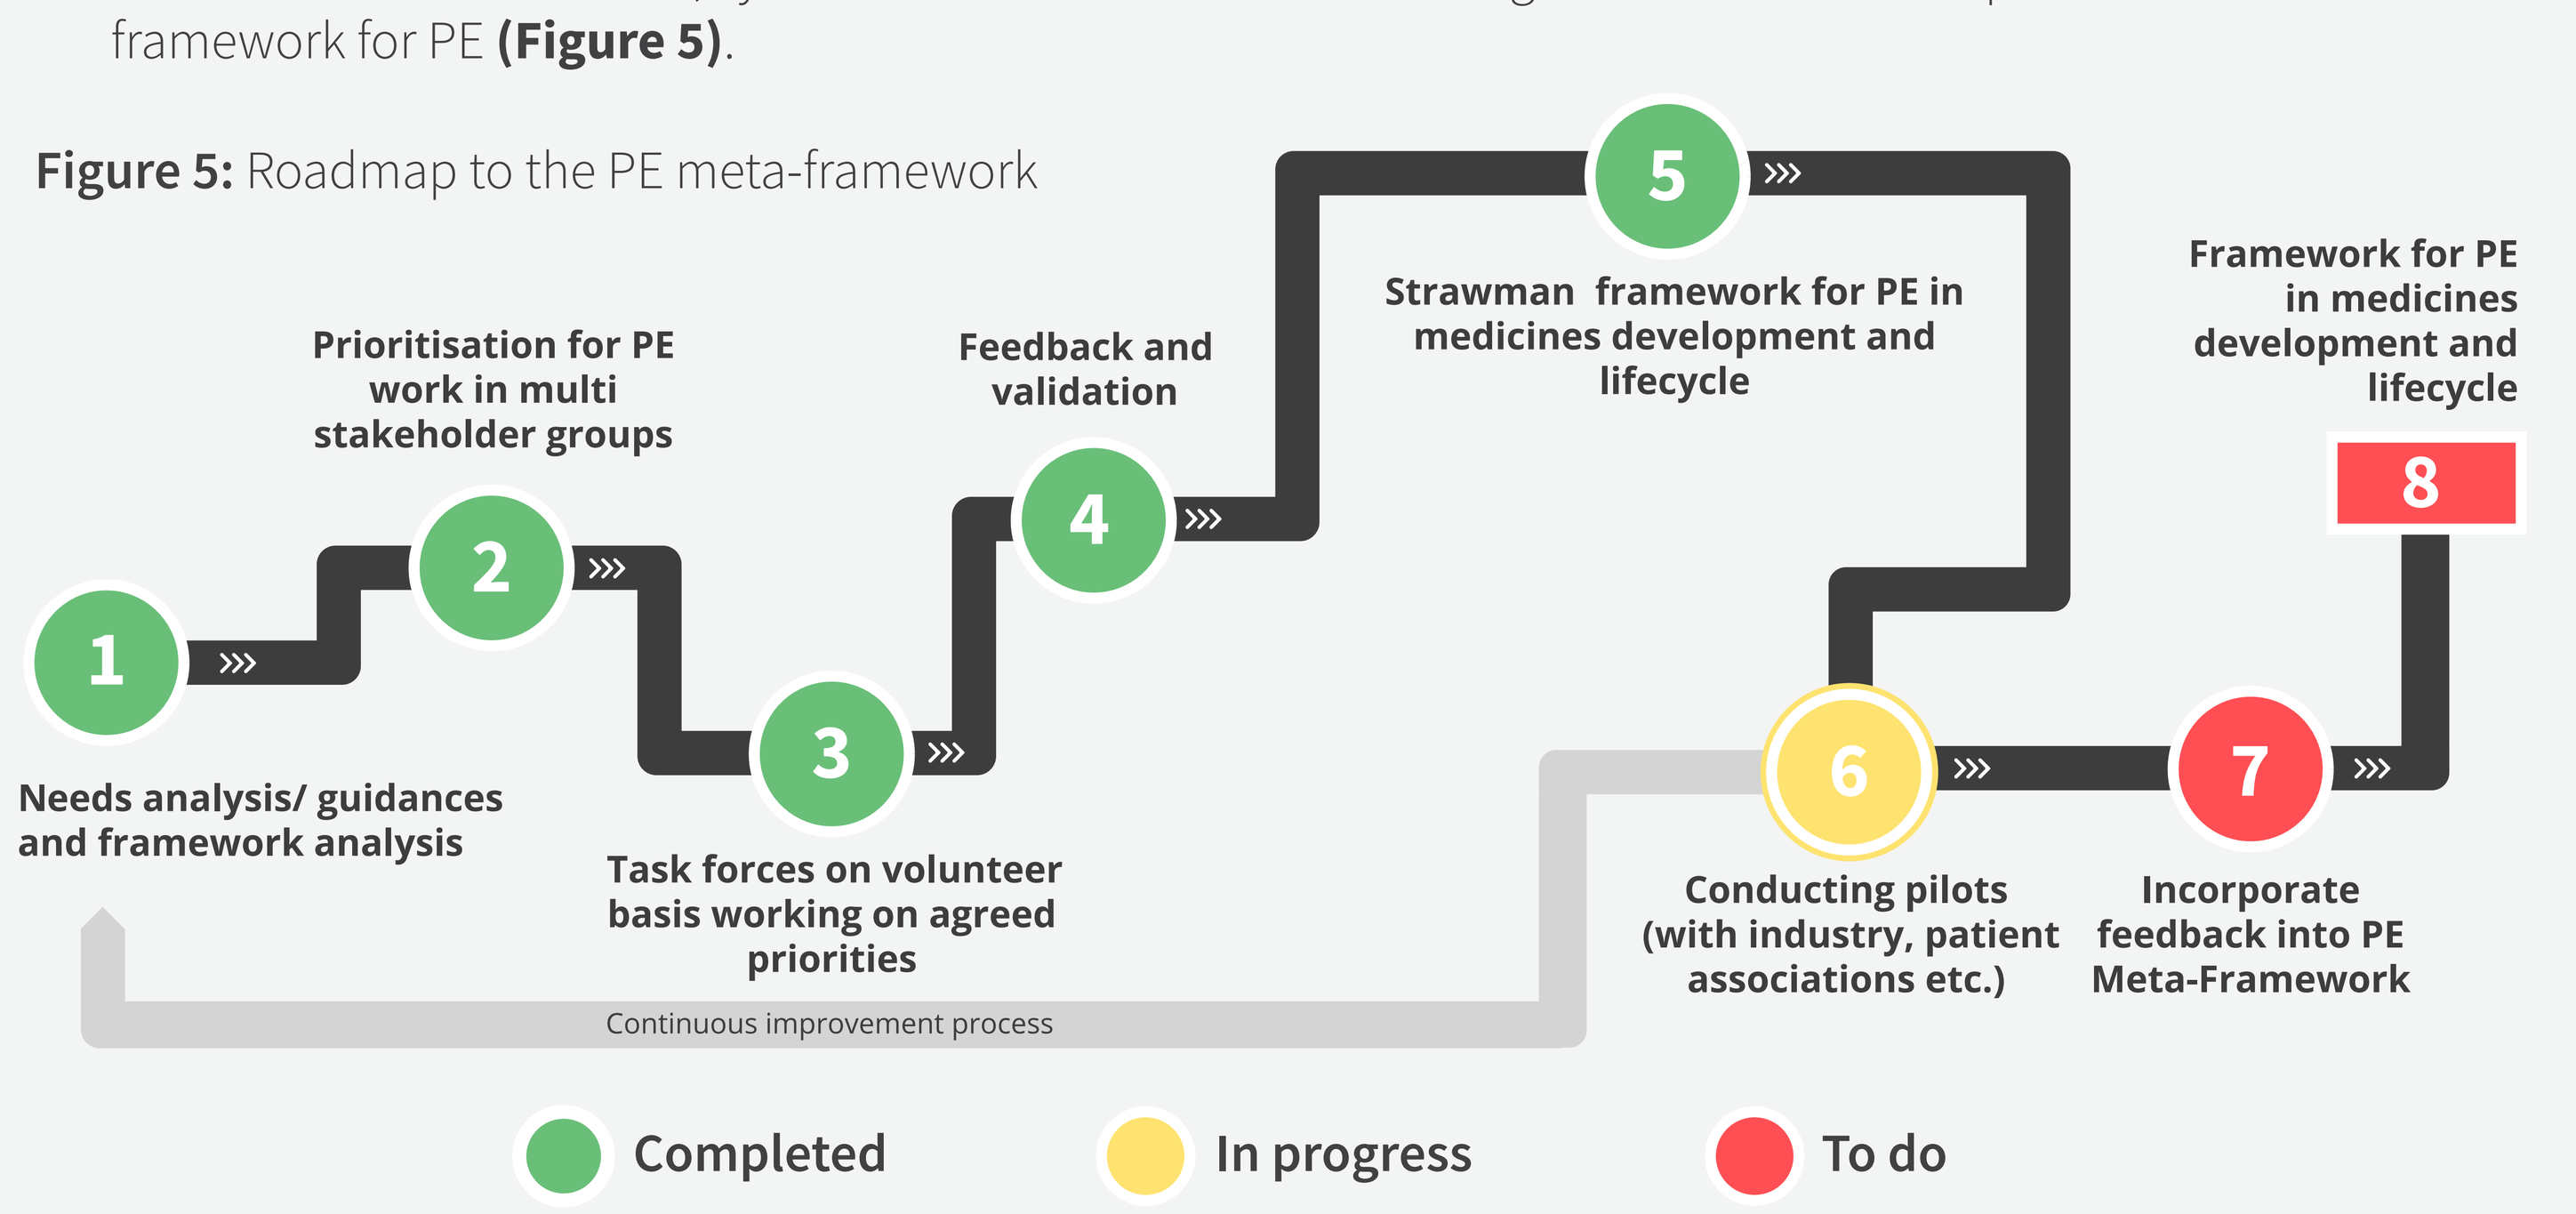
1 Needs analysis/ guidances and framework analysis
2 Prioritisation for PE work in multi stakeholder groups
3 Task forces on volunteer basis working on agreed priorities
4 Feedback and validation
5 Strawman framework for PE in medicines development and lifecycle
6 Conducting pilots (with industry, patient associations etc.)
7 Incorporate feedback into PE Meta-Framework
8 Framework for PE in medicines development and lifecycle
Continuous improvement process
Completed
In progress
To do

REFERENCES

1. CTTI (Clinical Trials Transformation Initiative) 2015. Available at: https://www.ctti-clinicaltrials.org/files/pgctrecs.pdf. Accessed September 23, 2018.
2. M-CERSI 2015. Available at: http://www.pharmacy.umaryland.edu/media/SOP/wwwpharmacyumarylandedu/centers/cersievents/pfdd/mcersi-pfdd-framework-rubric.pdf. Accessed September 23, 2018.
3. NHC/Genetic Alliance 2015. Available at: https://www.nationalhealthcouncil.org/sites/default/files/PatientEngagement-WhitePaper.pdf. Accessed September 23, 2018.
4. PCORI 2014a. PCORI Engagement Rubric for Applicants (Updated June 6, 2016). Available at: https://www.pcori.org/sites/default/files/Engagement-Rubric.pdf. Accessed September 23, 2018.
5. PCORI 2014b. Available at: https://www.pcori.org/sites/default/files/PCORI-Sample-Methods-Engagement-Plans.pdf. Accessed September 23, 2018.
6. Dubois RW, Lauer M, Perfetto E. Comp Eff Res 2013;2:383–91.
7. FasterCures 2016. Available at: http://www.fastercures.org/reports/view/56. Accessed September 23, 2018.
8. INVOLVE 2018a. Available at: http://www.invo.org.uk/wp-content/uploads/2018/06/Public_Involvement_Standards_v1.pdf. Accessed September 23, 2018.
9. INVOLVE 2018b. Available at http://www.invo.org.uk/wp-content/uploads/2018/03/Copro_Guidance_Mar18.pdf. Accessed September 23, 2018.
10. INVOLVE 2015. Available at: http://www.invo.org.uk/wp-content/uploads/2017/08/Values-Principles-framework-Jan2016.pdf. Accessed September 23, 2018.

GET POSTER PDF

Copies of this poster obtained through Quick Response Code are for personal use only and may not be reproduced without written permission of the authors.

GET PE QUALITY GUIDANCE TOOL

A practical guide to planning, developing and assessing the quality of patient engagement activities and projects throughout the development and lifecycle of medicines.

EXPLORE SYNaPsE

Patient engagement initiatives, organisations, experts and resources from around the world in one place